Real-World Treatment Patterns and Clinical Effectiveness of Palbociclib Plus an Aromatase Inhibitor as First-Line Therapy in Advanced/Metastatic Breast Cancer: Analysis From the Syapse Learning Health Network

Jeanna Wallenta Law,[1] Debanjali Mitra,[2] Henry Kaplan,[3] Tamuno Alfred,[2] Adam Brufsky,[4] Birol Emir,[2] Haley McCracken,[1] Xianchen Liu,[2] Ronda Broome,[1] Chenan Zhang,[1] Caroline DiCristo,[2] Connie Chen[2]

[1]Syapse, San Francisco, CA; [2]Pfizer Inc, New York, NY; [3]Swedish Cancer Institute, Seattle, WA; [4]Comprehensive Breast Cancer Center, University of Pittsburgh Medical Center, Pittsburgh, PA

Objective

- To describe real-world treatment patterns and clinical outcomes of patients with hormone receptor–positive/human epidermal growth factor receptor 2–negative (HR+/HER2–) advanced or metastatic breast cancer (A/MBC) who received palbociclib plus an aromatase inhibitor (AI) as first-line therapy in US practices.

Conclusion

- Real-world effectiveness outcomes in a heterogeneous population of patients with HR+/HER2– A/MBC receiving first-line palbociclib plus AI from the Syapse network of community oncologists demonstrated a median rwPFS of 31.7 months (95% CI, 26.5–50.3).
- These data complement randomized controlled trial data and other US electronic health record studies, supporting the continued use of palbociclib plus AI in the first-line setting.
- Limitations of this study include the single-arm nature of this analysis, a small sample size, and limited follow-up time.

Click or scan this quick response (QR) code to download this poster. Copies of this poster obtained through the QR Code are for personal use only and may not be reproduced without permission from the author of this poster. To request permission or to ask questions about the poster, please contact **Hank.Kaplan@swedish.org**.

References: 1. Howlader N, et al. *J Natl Cancer Inst.* 2014;106(5):5. **2.** Institute NC. SEER Stat Fact Sheets: Breast (Female). Available at: https://seer.cancer.gov/statfacts/html/breast.html. Accessed February 9, 2021. **3.** IBRANCE® (palbociclib). Full Prescribing Information, Pfizer Inc, New York, NY, 2019.

Disclosures: This study A5481155 was sponsored by Pfizer Inc. Editorial support was provided by Jill Shults, PhD, of ICON plc (North Wales, PA, USA) and was funded by Pfizer Inc.

Presented at the 38th Annual Miami Breast Cancer Conference • March 4–7, 2021 • Virtual Interactive Conference

Background

- Hormone receptor–positive/human epidermal growth factor receptor 2–negative (HR+/HER2–) breast cancer is the most common breast cancer subtype comprising >70% of breast cancers in the United States.[1]
- Women who have breast cancer with distant metastases have a poor prognosis, with a 5-year relative survival rate of 28%.[2]
- Palbociclib, a selective cyclin-dependent kinase 4/6 (CDK4/6) inhibitor, was approved in 2015 for use in postmenopausal women with HR+/HER2– advanced or metastatic breast cancer (A/MBC) in combination with an aromatase inhibitor (AI) as initial endocrine-based therapy.[3]
- There is now nearly 6 years of prescribing experience since the US approval of palbociclib.
- This retrospective single-arm study described real-world treatment patterns and clinical outcomes of patients with HR+/HER2– A/MBC who received palbociclib plus AI as first-line therapy in US practices.

Methods

- The study was a retrospective review of patient medical records that used the Syapse Learning Health Network, a longitudinal database that collects cancer care data from the entirety of a patient's journey from US health systems across 25 states, 457 hospitals, and 1317 community oncologists.
- Death data were captured using multiple sources, including the Surveillance, Epidemiology, and End Results (SEER) national cancer registry, other regional registries, the Social Security Death Index (SSDI), and digitized obituary data.
- Patients aged ≥18 years with HR+/HER2– A/MBC who initiated first-line palbociclib plus AI between February 3, 2015 and July 31, 2019 were included. Data cutoff was February 1, 2020, providing a minimum of 6 months of potential follow-up.

Key exclusion criteria:

- Enrolled in a clinical trial for A/MBC during the study observation period
- Evidence of prior treatment with any CDK4/6 inhibitor prior to the advanced setting
- Evidence of another primary cancer within 3 years before the initial line of therapy containing palbociclib

- Patients were followed from treatment initiation until data cutoff, death, or loss of contact, whichever came first.
- Treatment patterns and effectiveness outcomes were assessed.
 - Real-world progression-free survival (rwPFS) was defined as the time from the start of first-line therapy to a clinician-assessed progression event or death. Patients with no progression or death were censored at the earliest of the date of last contact or data cutoff.
 - Overall survival (OS) was defined as the time from the start of the first-line therapy to death. Patients not known to be deceased at the time of analysis were censored at the earliest of the date of last contact or data cutoff.
 - Real-world time to treatment discontinuation was defined as the time from the start of the first-line therapy to first-line treatment discontinuation or death. Patients with no discontinuation or death were censored at the earliest of last known usage of first-line treatment or data cutoff.
- Descriptive analyses were used for categorical variables. Kaplan-Meier survival analyses and landmark analyses were used to descriptively characterize time to event outcomes.

Results

- The median time from start of first-line treatment to the end of follow-up was 22.4 months.
- The study included 242 patients; baseline demographic characteristics are shown in **Table 1** and baseline clinical characteristics are shown in **Table 2**.
 - 50.8% of patients had bone-only disease and 32.2% had visceral disease.

Table 1. Baseline Demographic Characteristics (N=242)

Variable	**N (%)**
Race	
White	196 (81.0)
Black	29 (12.0)
Asian	10 (4.1)
Other or not provided	7 (2.9)
Ethnicity	
Hispanic-Latino	15 (6.2)
Non-Hispanic/Non-Latino	225 (93.0)
Unknown	2 (0.8)
Region of residence	
North central	230 (95.0)
South	10 (4.1)
Other	2 (0.8)
A/MBC diagnosis, y	
Median (min, max)	66.0 (31.0, 93.0)
<50	25 (10.3)
50–64	85 (35.1)
65–74	81 (33.5)
>75	51 (21.1)
Menopausal status at A/MBC diagnosis	
Postmenopausal	207 (85.5)
Premenopausal	24 (9.9)
Perimenopausal	2 (0.8)
Unknown/Not applicable	9 (3.7)
Insurance carrier	
Medicare/Medicaid	149 (61.6)
Commercial	69 (28.5)

A/MBC=advanced/metastatic breast cancer.

Table 2. Baseline Clinical Characteristics (N=242)

Variable	**N (%)**
Advanced/metastatic at diagnosis	137 (56.6)
Early stage at diagnosis	105 (43.4)
ECOG PS	
0	57 (23.6)
1	58 (24.0)
2+	29 (12.0)
Unknown	98 (40.5)
Comorbidities	
Hypertension	118 (48.8)
Diabetes	53 (21.9)
Renal disease	22 (9.1)
Chronic pulmonary disease	14 (5.8)
None[a]	89 (36.8)
DFI[b] in patients with early stage at diagnosis (N=105), mo	
≤12	14 (13.3)
13–24	12 (11.4)
25–36	10 (9.5)
>36	69 (65.7)
Number of metastatic sites	
0	1 (0.4)
1	147 (60.7)
2	45 (18.6)
>3	49 (20.2)
Sites of metastases[c]	
Bone (bone only or in addition to other sites)	201 (83.1)
Bone only	123 (50.8)
Visceral	78 (32.2)
Lung	52 (21.5)
Distant lymph nodes	45 (18.6)
Liver	24 (9.9)
Malignant pleural effusion	25 (10.3)
Primary endocrine resistance	
Yes	69 (28.5)
No	173 (71.5)

DFI=disease-free interval; ECOG PS=Eastern Cooperative Oncology Group Performance Status.
[a]None of the searched comorbidities which included: anemia, heart arrhythmia, hypertension, myocardial infarction, congestive heart failure, peripheral vascular disease, cerebrovascular accident, hemiplegia, chronic obstructive pulmonary disease, ulcer disease, diabetes with chronic complications, diabetes without chronic complications, renal disease, connective tissue disease such as rheumatoid arthritis or lupus, Alzheimer's or other dementia, cirrhosis or other serious liver disease, mild liver disease, non-breast cancer malignancy (excluding sites of breast cancer metastasis and malignant neoplasm of skin), metastatic solid tumor (other than breast cancer), human immunodeficiency virus infection/acquired immune deficiency syndrome.
[b]Defined as from the end of adjuvant treatment to disease recurrence (months).
[c]Metastatic sites occurring in ≥9.9% of patients.

Figure 1. Kaplan-Meier Analysis of Real-World Progression-Free Survival

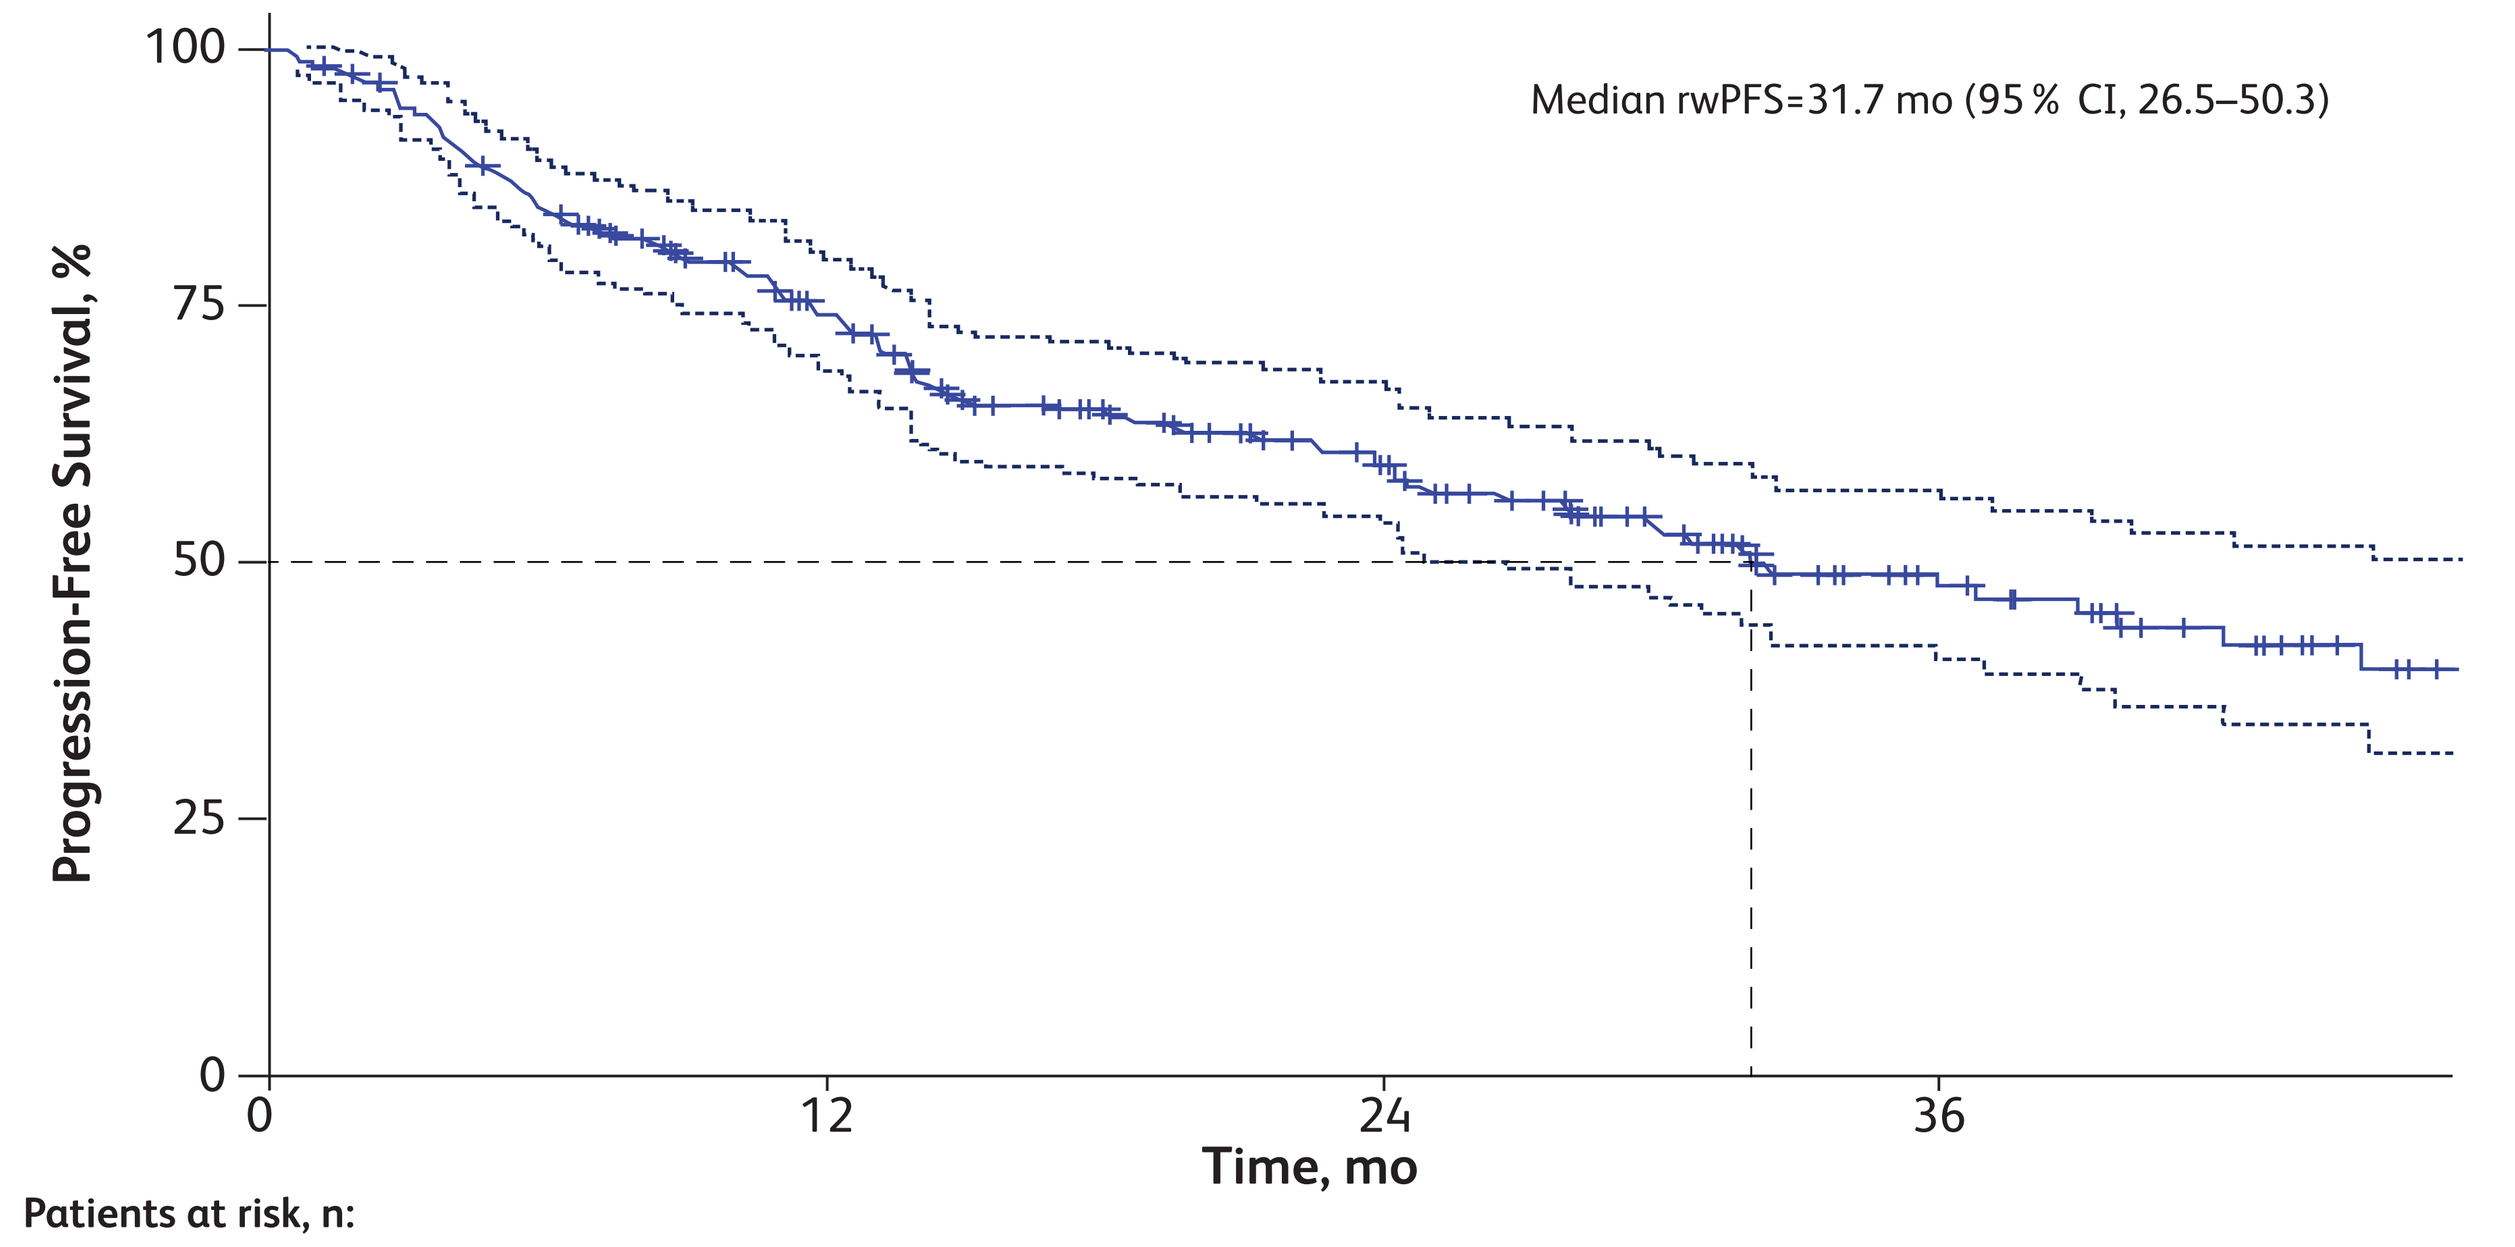
Median rwPFS=31.7 mo (95 % CI, 26.5–50.3)
Progression-Free Survival, %
100
75
50
25
0
0
12
24
36
Time, mo

	0	12	24	36
Patients at risk, n:	242	155	89	39
Cumlative number of events:	0	60	88	102

rwPFS Rate, %					
6 mo	12 mo	18 mo	24 mo	30 mo	36 mo
84	74	64	59	53	48

rwPFS=real-world progression-free survival.

Figure 2. Kaplan-Meier Analysis of Real-World Time to Treatment Discontinuation

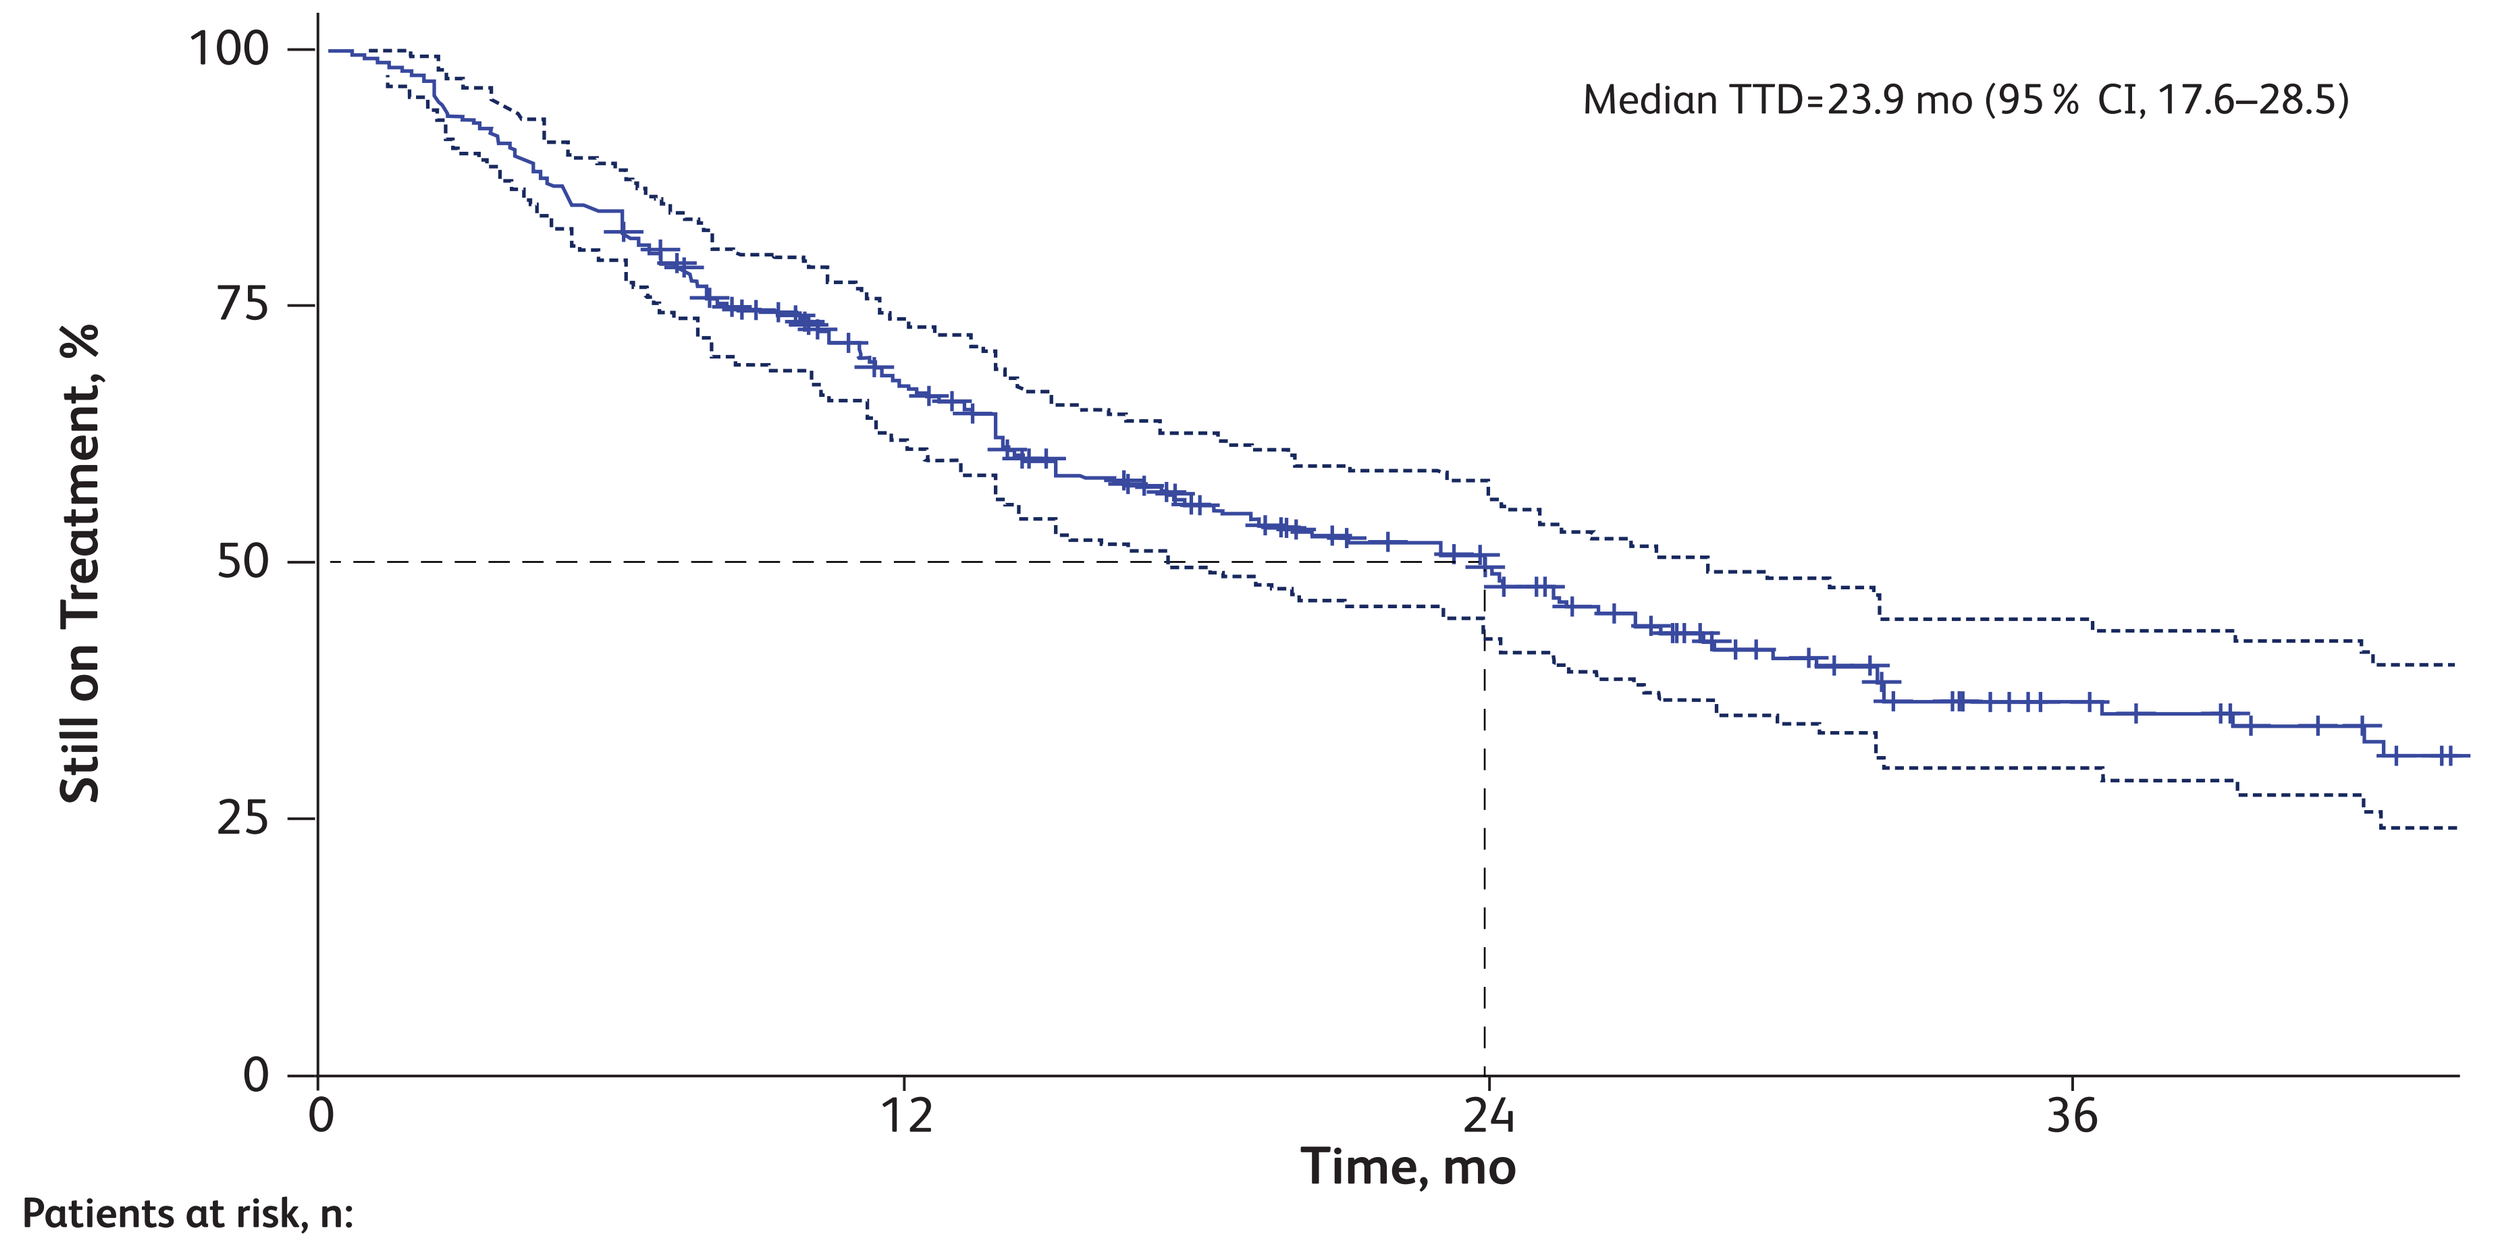
Median TTD=23.9 mo (95 % CI, 17.6–28.5)
Still on Treatment, %
100
75
50
25
0
0
12
24
36
Time, mo

	0	12	24	36
Patients at risk, n:	242	145	78	33
Cumlative number of events:	0	77	113	130

Still on Treatment Rate, %					
6 mo	12 mo	18 mo	24 mo	30 mo	36 mo
84	67	56	49	41	36

TTD=time to treatment discontinuation.

- The most common reasons for treatment discontinuation were progressive disease (26%) and intolerance/toxicity (15%); 44% of patients remained on treatment at study cutoff.
- Of patients with a known dose adjustment (31%), median time to adjustment was 55 days.
- Median rwPFS was 31.7 months (95% CI, 26.5–50.3; **Figure 1**).
- OS rates were 90% (95% CI, 87–94) and 78% (95% CI, 72–84) at 12 months and 24 months, respectively; 25.6% of patients died during the study.
- Median time to treatment discontinuation was 23.9 months (95% CI, 17.6–28.5; **Figure 2**).

LIMITATIONS

- This study is limited by a small sample size and limited follow-up to reliably estimate median OS.
- With retrospective studies, there is the potential for missing data that were unable to be collected.

Copyright ©2021. All rights reserved.